1218

In Vitro Activity of Lefamulin against *S. aureus* Collected Worldwide from Hospitalized Patients with Bacterial Pneumonia

Paukner, Susanne [1]; Flamm, Robert K. [2]; Schuchert, Jason [2]; Gelone, Steven P. [3]; Sader, Helio S. [2]

[1] Nabriva Therapeutics, Vienna, Austria; [2] JMI Laboratories, North Liberty, IA, USA; [3] Nabriva Therapeutics, King of Prussia, PA, USA

Nabriva Therapeutics
www.nabriva.com
susanne.paukner@nabriva.com

ABSTRACT

Background: *S. aureus* (SA) is a well-recognized cause of pneumonia from both the community and hospital settings.[1] The clinical management of SA pneumonia is complicated by the invasive infection it can cause and the high prevalence of methicillin resistance (MR).[2] Lefamulin (LEF) is the first semi-synthetic pleuromutilin antibiotic for IV and oral use in humans and it specifically inhibits bacterial protein synthesis. LEF is currently in Phase 3 trials for the treatment of community-acquired bacterial pneumonia (CABP). This study investigated the *in vitro* activity of LEF and comparators against SA strains collected from patients hospitalized with pneumonia in 2015.

Methods: 1,273 unique SA isolates were collected from hospitalized patients with pneumonia worldwide in 28 countries (33 sites) in 2015 as part of the SENTRY surveillance program. Isolates included 401 hospital-acquired (HA) SA (259 from ICU, 152 from ventilator associated pneumonia, [VAP]). Susceptibility testing was conducted using the CLSI broth microdilution method and susceptibility was interpreted per CLSI 2017 breakpoint criteria.

Results: LEF was the most potent compound tested, with 99.7% of all SA isolates being inhibited at a concentration of ≤0.25 mg/L ($MIC_{50/90}$ values of 0.06/0.12 mg/L) and irrespective of the collection source (ICU/non-ICU, VAP/non-VAP). 31.6% of isolates (*n*=402) were MRSA of which 99.3% were inhibited at a LEF concentration of ≤0.25 µg/mL ($MIC_{50/90}$, 0.06/0.12 mg/L). Susceptibility rates for all SA isolates were >90% for ceftaroline, vancomycin, linezolid and doxycycline. Susceptibility to azithromycin, levofloxacin and clindamycin was limited, particularly among MRSA (see Table 1).

Conclusion: SA strains collected from patients hospitalized with pneumonia including HAP and VAP were highly susceptible to LEF regardless of the resistance phenotype to the other antibiotics tested. Due to its potent activity against resistant SA and the most prevalent typical and atypical respiratory pathogens, as well as the availability of IV and oral formulations, LEF has the potential to play a role in the empiric treatment of CABP and supports evaluation in HAP and VAP caused by SA.

Table 1. *In vitro* activity of lefamulin and comparators [mg/L]

Compound	*S. aureus*, total (*n*=1273)		MSSA (*n*=871)		MRSA (*n*=402)	
	MIC_{50}	MIC_{90}	% S	% R	% S	% R
Lefamulin	0.06	0.12	-	-	-	-
Azithromycin	0.5	>4	73.9	23.9	19.2	79.9
Ceftaroline	0.25	1	100.0	0.0	78.9	0.2
Clindamycin	≤0.25	>2	100.0	0.0	57.0	43.0
Doxycycline	≤0.06	0.25	99.4	0.1	93.8	0.2
Levofloxacin	0.25	>4	93.8	5.5	22.4	77.1
Oxacillin	0.5	>2	100.0	0.0	0.0	100.0
Linezolid	1	2	99.9	0.1	100.0	0.0
Vancomycin	0.5	1	100.0	0.0	100.0	0.0

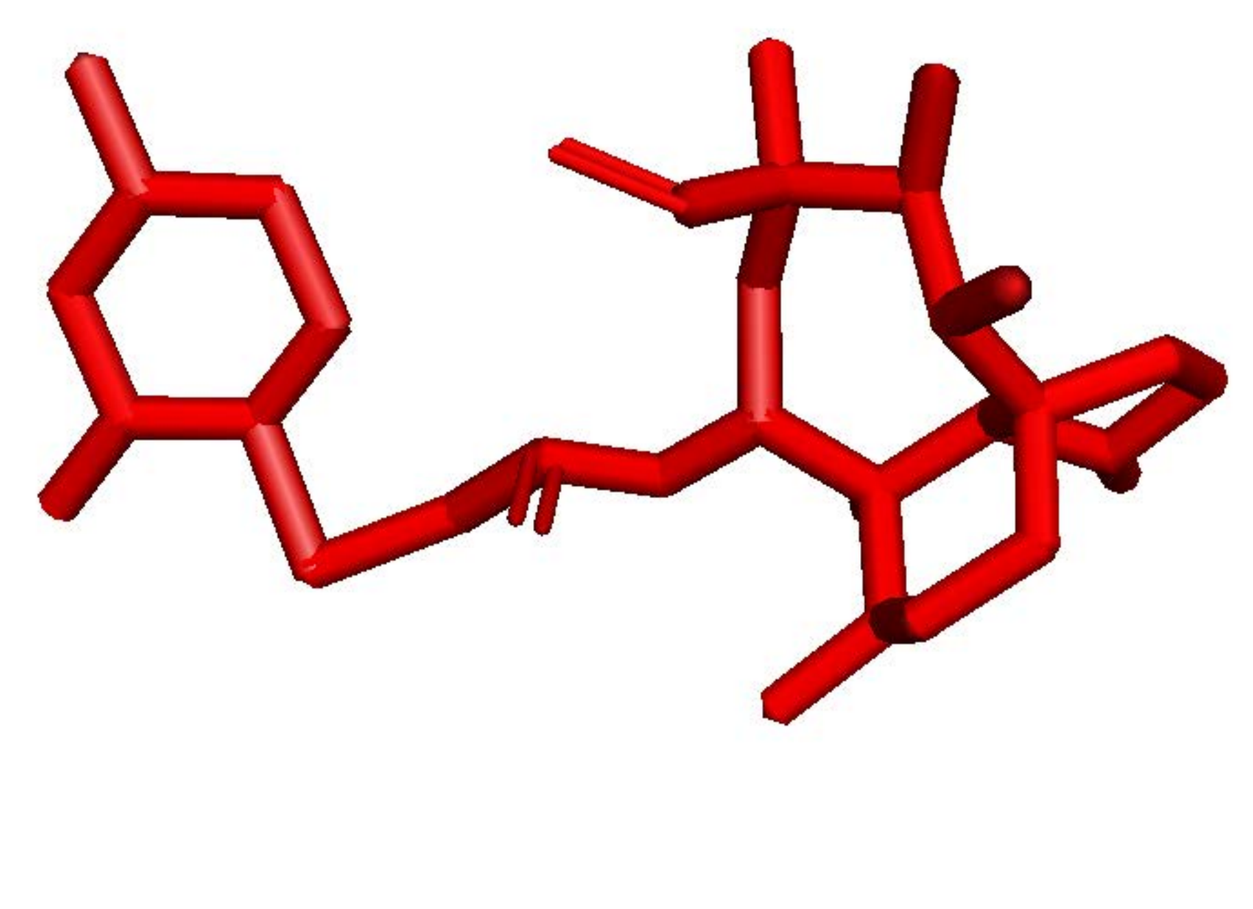

Figure 1. Lefamulin

INTRODUCTION

Lefamulin is the first representative of pleuromutilin class in clinical development for systemic administration. Pleuromutilins inhibit bacterial protein synthesis of Gram-positive and Gram-negative organisms, as well as atypical respiratory pathogens.[3,4] Lefamulin effectively and selectively inhibits bacterial translation by binding to the peptidyl transferase center (PTC) via four H-bonds and other interactions at the A- and P-sites, resulting in an "induced fit."[5,6] Phase 1 and 2 trials have demonstrated that IV and oral administration of lefamulin are well tolerated. Furthermore, lefamulin (100 mg or 150 mg IV q12 hours) showed similar efficacy to IV vancomycin in a clinical Phase 2 trial in patients with acute bacterial skin and skin structure infections (ABSSSI).[7] Currently lefamulin is in late stage development for the treatment of community-acquired bacterial pneumonia (CABP).

RESULTS

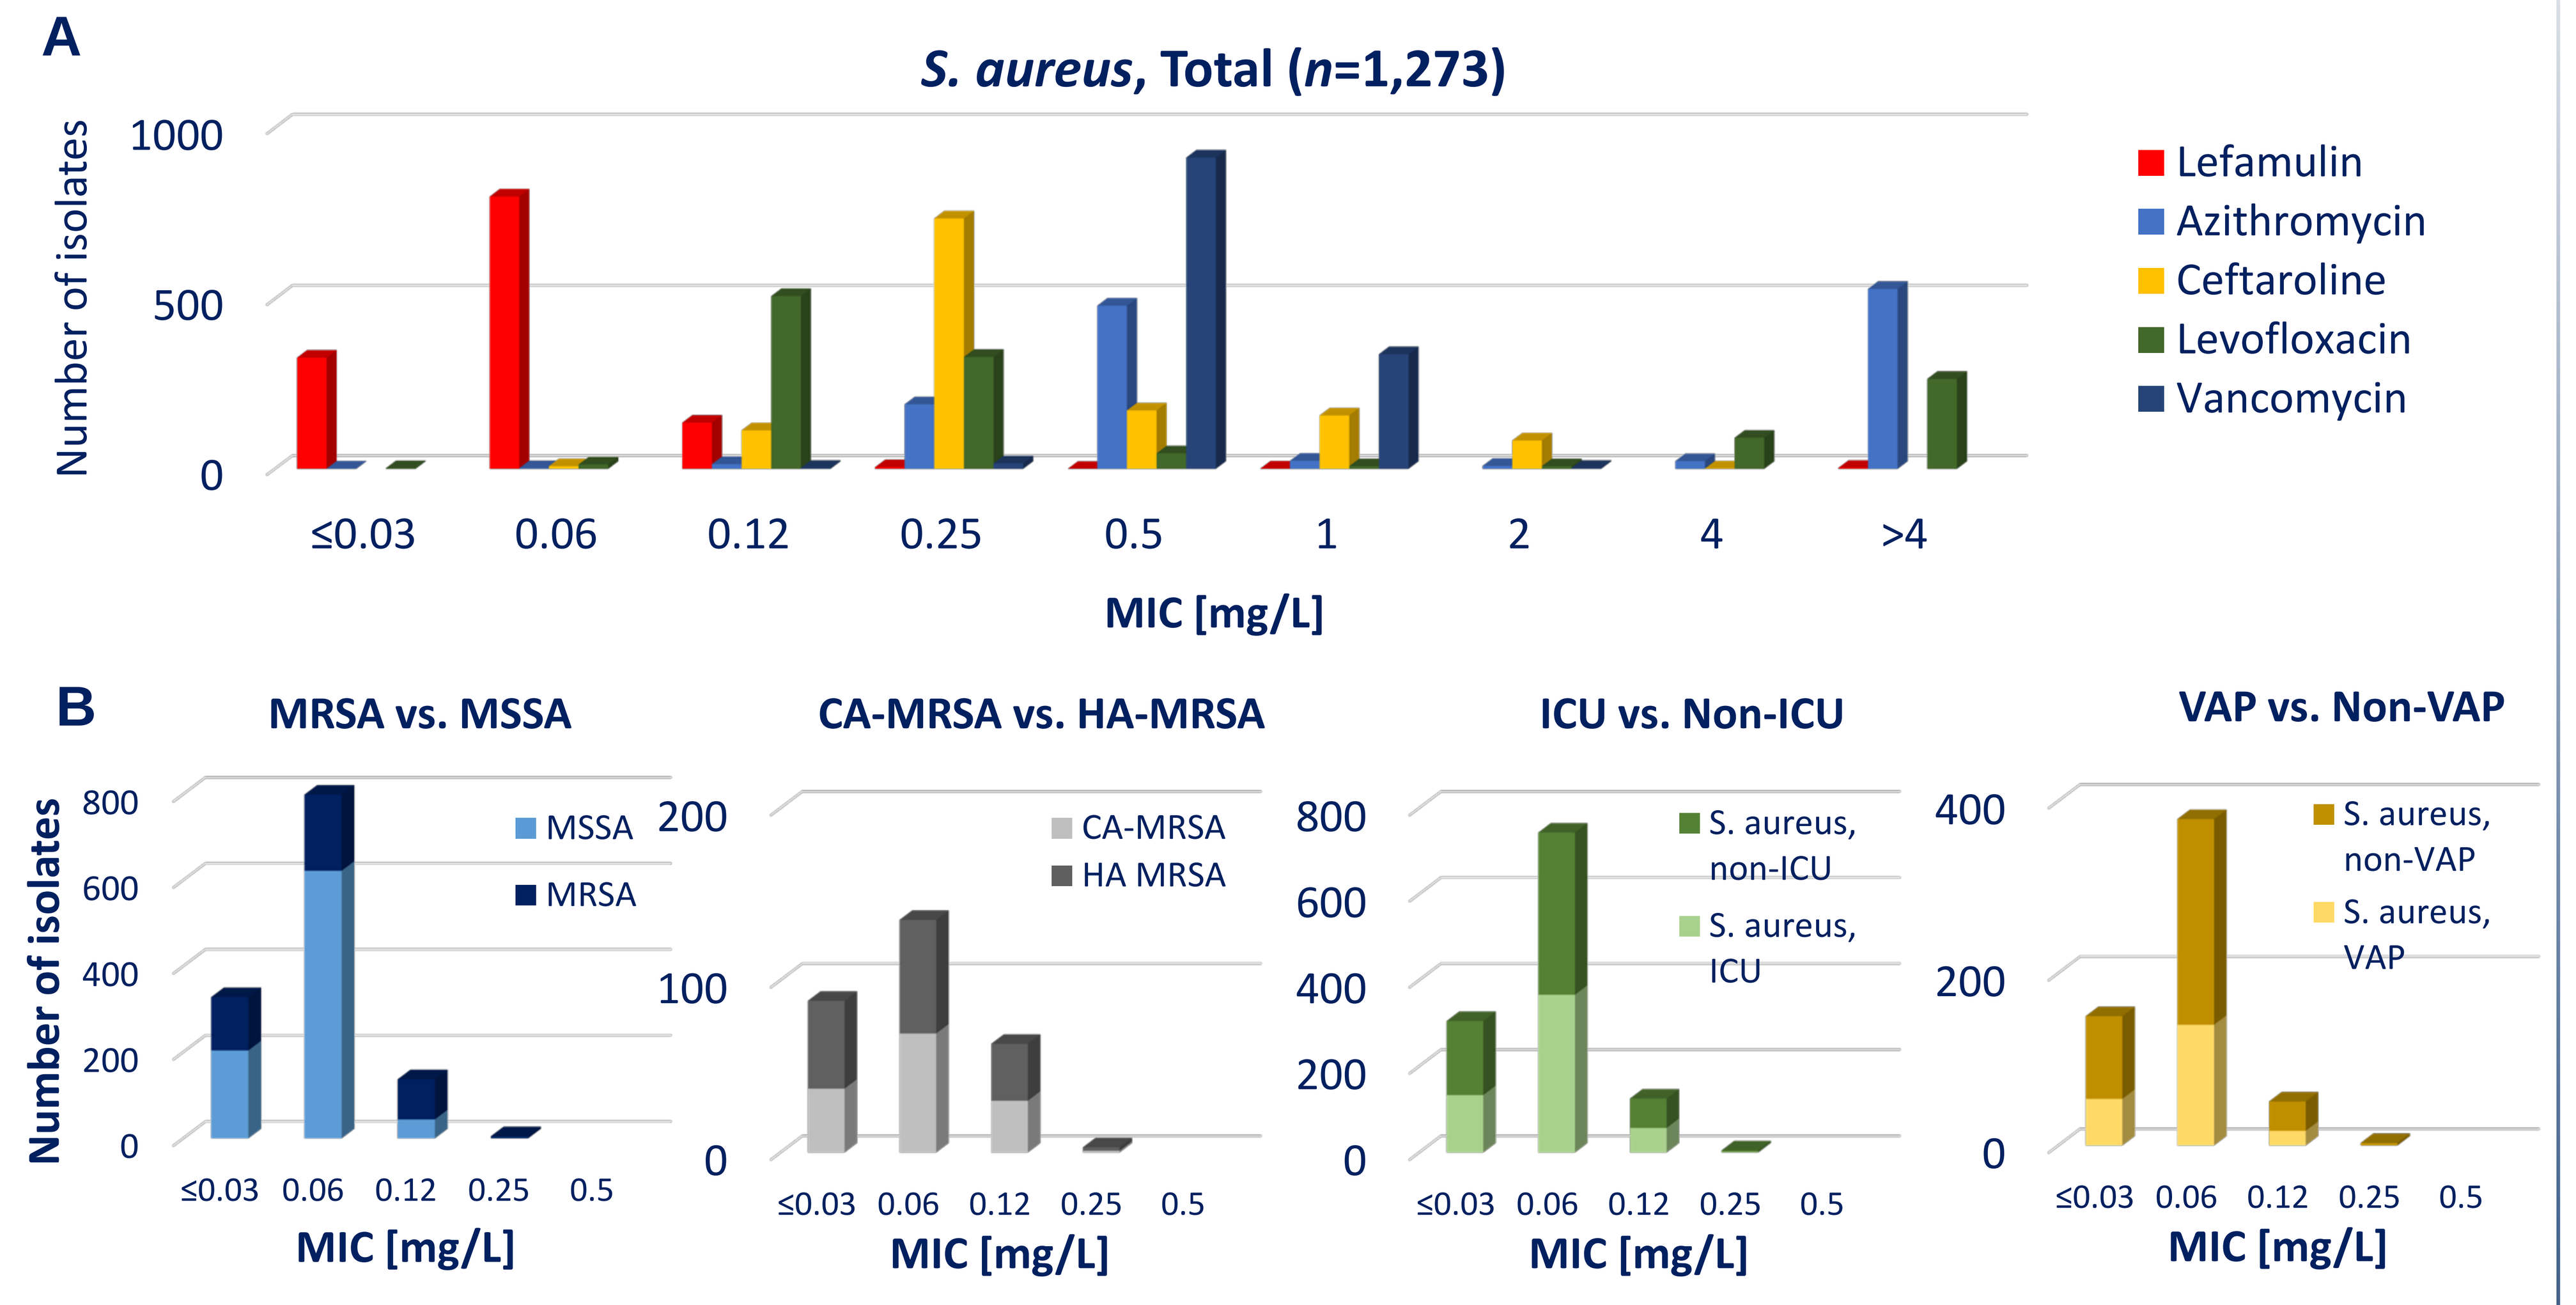
A
S. aureus, Total (*n*=1,273)
Number of isolates
1000
500
0
≤0.03
0.06
0.12
0.25
0.5
1
2
4
>4
MIC [mg/L]
Lefamulin
Azithromycin
Ceftaroline
Levofloxacin
Vancomycin
B
MRSA vs. MSSA
CA-MRSA vs. HA-MRSA
ICU vs. Non-ICU
VAP vs. Non-VAP
Number of isolates
MSSA
MRSA
CA-MRSA
HA MRSA
S. aureus, non-ICU
S. aureus, ICU
S. aureus, non-VAP
S. aureus, VAP
MIC [mg/L]

Figure 2. MIC distributions of lefamulin and comparators (A) and lefamulin MIC distribution for various *S. aureus* subsets (B)

Table 2. *In vitro* activity of lefamulin and comparators [mg/L]

Organism (*n*)	MIC_{50}	MIC_{90}	MIC_{99}	Range [mg/L]	% S [a]	% I [a]	% R [a]
***S. aureus* (1,273)**							
Lefamulin	0.06	0.12	0.12	≤0.03 - >1 [b]	-	-	-
Azithromycin	0.5	>4	>4	≤0.03 - >4	56.6	1.8	41.6
Ceftaroline	0.25	1	2	≤0.06 - 4	93.3	6.6	0.1
Clindamycin	≤0.25	>2	>2	≤0.25 - >2	83.9	0.2	15.9
Doxycycline	≤0.06	0.25	8	≤0.06 - >8	97.6	2.2	0.2
Erythromycin	0.25	>8	>8	≤0.06 - >8	56.2	5.1	38.6
Levofloxacin	0.25	>4	>4	≤0.03 - >4	71.2	0.6	28.1
Linezolid	1	1	2	0.25 - 8	99.9	-	0.1
Oxacillin	0.5	>2	>2	≤0.25 - >2	68.4	-	31.6
Vancomycin	0.5	1	1	≤0.12 - 2	100.0	0.0	0.0
MRSA (402)							
Lefamulin	0.06	0.12	0.25	≤0.03 - >1			
Azithromycin	>4	>4	>4	0.06 - >4	19.2	1.0	79.9
Ceftaroline	1	2	2	0.25 - 4	78.9	20.9	0.2
Clindamycin	≤0.25	>2	>2	≤0.25 - >2	57.0	0.0	43.0
Doxycycline	≤0.06	4	8	≤0.06 - >8	93.8	6.0	0.2
Erythromycin	>8	>8	>8	≤0.06 - >8	18.9	4.7	76.4
Levofloxacin	>4	>4	>4	0.06 - >4	22.4	0.5	77.1
Linezolid	1	1	2	0.25 - 2	100.0	-	0.0
Oxacillin	>2	>2	>2	>2 - >2	0.0	-	100.0
Vancomycin	0.5	1	1	≤0.12 - 2	100.0	0.0	0.0

[a] Criteria as published by CLSI [2017]; [b] Two of 1,273 isolates showed a lefamulin MIC of >1 mg/L.

- Lefamulin showed potent antibacterial activity against *S. aureus* isolates, including resistant isolates (Figure 2, Tables 1 and 2).
- Lefamulin was the most potent compound tested with 99.3% inhibited at a lefamulin concentration of ≤0.25 mg/L ($MIC_{50/90}$ values of 0.06/0.12 mg/L).
- The lefamulin activity was not affected by resistance to other antibiotics tested, including macrolides, tetracyclines, fluoroquinolones, or ß-lactam antibiotics or the nosocomial status of isolates.
 - 31.6% of isolates were MRSA, of which *n*=139 community-acquired (CA-MRSA) and *n*=152 were nosocomial (HA-MRSA).
- Lefamulin MIC distributions were similar between subsets, including MSSA vs. MRSA, community-acquired vs. nosocomial isolates, ICU vs. non-ICU and VAP vs. non-VAP (Figure 2B).
 - MSSA and MRSA showed lefamulin $MIC_{50/90}$ of 0.06/0.12 mg/L.
 - Both CA-SA (*n*=528; MSSA and MRSA) and HA-SA (*n*=401) displayed a lefamulin $MIC_{50/90}$ of 0.06/0.12 mg/L $MIC_{50/90}$.
- *S. aureus* isolates were largely susceptible to doxycycline (97.6%), ceftaroline (93.3%), linezolid (99.9%) and vancomycin (100%).
- Susceptibility to macrolides, fluoroquinolones and lincosamides was significantly reduced, particularly among MRSA.
 - 79.9% of MRSA and 23.9% of MSSA were resistant to azithromycin.
 - 77.1% of MRSA and 5.5% of MSSA were resistant to levofloxacin.
 - 43.0% of MRSA and 3.4% of MSSA were resistant to clindamycin.

CONCLUSIONS

- **Lefamulin displayed potent *in vitro* activity against this contemporary collection of *S. aureus* isolates collected from hospitalized patients with pneumonia.**
- **Lefamulin was the most active compound against MRSA and MSSA, including both, community-acquired and nosocomial isolates, irrespective of the resistance phenotype to other antibiotic classes including macrolides, fluoroquinolones, tetracyclines or ß-lactam antibiotics.**
- **These data support the development of lefamulin for infections caused by *S. aureus*, including CAP, HAP and ABSSSI.**

REFERENCES

(1) Jain S., et al. *NEJM* 373, 415-27 (2015)
(2) http://ecdc.europa.eu/en/healthtopics/Health care-associated_infections/guidance-infection-preventioncontrol/Pages/guidance-prevention-control-infections-MRSA.aspx
(3) Paukner, et al. *AAC* 57(9), 4489-4495 (2013)
(4) Waites, K. B., et al. *AAC* 61(2)(2017)
(5) Eyal, Z., et al., *Sci Rep* 6, 39004 (2016)
(6) Paukner, S. and Riedl, R. Cold Spring Harbor Laboratory Perspectives, *Antibiotics and Antibiotic Resistance* (2016)
(7) Prince, W. T, et al. *AAC* 57(5), 2087-2094 (2013)
(8) CLSI, M100(2017)

ID Week 2017, October 4-8, San Diego, CA, USA